Poster Number
W1800

Digestive Disease Week 2009 • McCormick Place, Chicago Illinois • May 30-June 4, 2009

Association between HBV Replication and Serum ALT Changes is not Dependent on HBV Genotype and Mutants

Academia Sinica
Genomics Research Center

Hwai-I Yang[1,2], Uchenna H Iloeje[3], Jun Su[3], Chin-Lan Jen[1,2], San-Lin You[1,2] and Chien-Jen Chen[1,2] for the R.E.V.E.A.L.-HBV Study Group

[1]Genomics Research Center, Academia Sinica, Taipei, Taiwan; [2]Graduate Institute of Epidemiology, College of Public Health, National Taiwan University, Taipei, Taiwan; [3]Research and Development, Bristol-Myers Squibb Company, Wallingford, USA

Background

- We previously demonstrated in a model-based trajectory analysis that elevated serum HBV DNA level predicted future changes in ALT level, which has been shown to be an important risk predictor of disease progression for patients with chronic hepatitis B[1]
- HBV genotype and specific mutations were not taken into consideration in the previous analysis
- We aimed to explore the impact of these new variables on the established association

Study population

- Source data: the R.E.V.E.A.L.-HBV study cohort (N=3,653)
 - Population-based prospective study with a mean follow-up of 11 years
 - Subjects aged 30–65 years
 - At study entry, subjects: 1) provided informed consent; 2) provided a serum sample which was separated on the day of collection and frozen at –70ºC; 3) underwent a structured interview by a trained public health nurse; 4) had an ultrasound examination
 - HBsAg-positive and anti-HCV-negative
- This analysis used a subset from the R.E.V.E.A.L.-HBV study cohort who had normal serum ALT level at baseline (<45 U/L) and had at least three ALT tests during follow-up (n=854)

Methods

- The outcome of interest: serum ALT elevation (≥45 U/L) during follow-up on at least one occasion
- Laboratory tests
 - HBsAg and HBeAg: radioimmunoassay (Abbott Laboratories, North Chicago, IL, USA)
 - Anti-HCV antibodies: second-generation ELISA kits (Abbott Laboratories, North Chicago, IL, USA)
 - ALT: serum chemistry autoanalyser (Model 736, Hitachi Co., Tokyo, Japan) using commercial reagents (bioMérieux, Mercy-l'Etoile, France)
 - Serum HBV DNA level: commercial PCR kits (Cobas Amplicor, Roche Diagnostics Co., Indianapolis, IN, USA)
 - HBV genotype: melting curve analysis
 - Precore and basal core promoter (BCP) mutations: direct sequencing
- Multiple logistic regression was used to derive multivariate-adjusted odds ratio (ORadj) and 95% confidence interval (CI) for risk factors associated with the outcome

Results

Table 1. Baseline Demographic and Disease Characteristics for All Subjects

	No ALT elevation during follow-up (n=534)	ALT elevation during follow-up (n=320)	p-value
Male, n (%)	388 (72.7)	270 (84.4)	<0.0001
Age, years, mean (median)	45.7 (45.0)	44.7 (44.0)	0.11
HBeAg-positive, n (%)	103 (19.3)	119 (37.2)	<0.0001
Serum ALT level, U/L			<0.0001
<15, n (%)	341 (63.9)	148 (46.3)	
15–44, n (%)	193 (36.1)	172 (53.8)	
Cigarette smoking yes, n (%)	206 (38.6)	126 (39.4)	0.82
Alcohol consumption yes, n (%)	65 (12.2)	45 (14.1)	0.42
HBV DNA level, copies/mL			<0.0001
<300, n (%)	47 (8.8)	10 (3.1)	
300–9,999, n (%)	152 (28.5)	24 (7.5)	
10,000–99,999, n (%)	138 (25.8)	61 (19.1)	
100,000–999,999, n (%)	83 (15.5)	86 (26.9)	
≥1,000,000, n (%)	114 (21.4)	139 (43.4)	
HBV genotype			0.14
B, n (%)	319 (59.7)	176 (55.0)	
C, n (%)	196 (36.7)	137 (42.8)	
B+C, n (%)	19 (3.6)	7 (2.2)	
Precore 1896			<0.0001
Wild type, n (%)	175 (32.8)	149 (46.6)	
G1896A, n (%)	316 (59.2)	137 (42.8)	
Mixed type, n (%)	43 (8.1)	34 (10.6)	
BCP 1762/1764			0.05
Wild type, n (%)	314 (58.8)	161 (50.3)	
A1762T/G1764A, n (%)	147 (27.5)	103 (32.2)	
Mixed type, n (%)	73 (13.7)	56 (17.5)	

Table 2. Univariate Analyses of the Risk of ALT Elevation During Follow-up

Variable	Crude odds ratio (95% CI)	p-value
Gender		
Female	1.0 (referent)	
Male	2.0 (1.4–2.9)	<0.0001
Age (1-year increment)	0.99 (0.97–1.0)	0.11
Cigarette smoking		
No	1.0 (referent)	
Yes	1.0 (0.8–1.4)	0.82
Alcohol consumption		
No	1.0 (referent)	
Yes	1.2 (0.8–1.8)	0.43
HBeAg		
Positive	1.0 (referent)	
Negative	2.5 (1.8–3.4)	<0.0001
Baseline HBV DNA level, copies/mL		
<300 (undetectable)	1.0 (referent)	
300–9,999	0.7 (0.3–1.7)	0.47
10,000–99,999	2.1 (1.0–4.4)	0.05
100,000–999,999	4.9 (2.3–10.3)	<0.0001
≥1,000,000	5.7 (2.8–11.8)	<0.0001
Trend test		<0.0001
Baseline serum ALT level, U/L		
<15	1.0 (referent)	
15–44	2.1 (1.5–2.7)	<0.0001
HBV genotype		
B	1.0 (referent)	
C	1.3 (0.9–1.7)	0.10
B+C	0.7 (0.3–1.6)	0.37
Precore 1896		
Wild type	1.0 (referent)	
G1896A	0.5 (0.4–0.7)	<0.0001
Mixed type	0.9 (0.6–1.5)	0.77
BCP 1762/1764		
Wild type	1.0 (referent)	
A1762T/G1764A	1.4 (1.0–1.9)	0.05
Mixed type	1.5 (1.0–2.2)	0.05

Table 3. Multivariate Logistic Regression Analysis of the Risk of ALT Elevation During Follow-up

Variable	Adjusted odds ratio (95% CI)	p-value
Gender		
Female	1.0 (referent)	
Male	2.6 (1.7–4.1)	<0.0001
Age (1-year increment)	0.99 (0.98–1.01)	0.41
Cigarette smoking		
No	1.0 (referent)	
Yes	0.8 (0.5–1.1)	0.09
Alcohol consumption		
No	1.0 (referent)	
Yes	0.9 (0.6–1.4)	0.63
HBeAg		
Positive	1.0 (referent)	
Negative	0.7 (0.4–1.3)	0.28
Baseline HBV DNA level, copies/mL		
<300 (undetectable)	1.0 (referent)	
300–9,999	0.8 (0.3–1.8)	0.53
10,000–99,999	2.3 (1.1–4.9)	0.037
100,000–999,999	4.6 (2.1–10.0)	0.0001
≥1,000,000	5.5 (2.4–12.9)	<0.0001
Trend test		<0.0001
Baseline serum ALT level, U/L		
<15	1.0 (referent)	
15–44	1.4 (1.0–1.9)	0.042
HBV genotype		
B	1.0 (referent)	
C	1.3 (0.9–1.9)	0.22
B+C	0.7 (0.3–1.7)	0.38
Precore 1896		
Wild type	1.0 (referent)	
G1896A	0.7 (0.5–1.1)	0.09
Mixed type	1.1 (0.6–2.0)	0.70
BCP 1762/1764		
Wild type	1.0 (referent)	
A1762T/G1764A	1.3 (0.9–1.8)	0.20
Mixed type	1.3 (0.8–2.1)	0.24

Table 4. Association between Baseline HBV Viral Load and ALT Elevation Stratified by Genotype

Baseline HBV DNA level copies/mL	Genotype B	Genotype C
	Adjusted odds ratio (95% CI)	Adjusted odds ratio (95% CI)
<300 (undetectable)	1.0 (referent)	1.0 (referent)
300–9,999	1.1 (0.3–3.8)	0.6 (0.2–2.2)
10,000–99,999	2.6 (0.8–8.1)	2.0 (0.6–7.1)
100,000–999,999	4.4 (1.4–13.8)*	6.0 (1.6–22.1)†
≥1,000,000	3.4 (1.0–11.8)	17.3 (4.2–71.6)‡
Trend test	<0.0001	<0.0001

Adjustment for gender, age, habits of cigarette smoking and alcohol consumption, HBeAg status and baseline ALT level
* $p<0.05$; † $p<0.01$; ‡ $p<0.001$.

Table 5. Association between Baseline HBV Viral Load and ALT Elevation Afterward with Stratification by Precore 1896 Variant

Baseline HBV DNA level copies/mL	Precore wild type	Precore G1896A
	Adjusted odds ratio (95% CI)	Adjusted odds ratio (95% CI)
<300 (undetectable)	1.0 (referent)	1.0 (referent)
300–9,999	2.0 (0.4–10.9)	0.4 (0.1–1.2)
10,000–99,999	6.3 (1.2–31.8)*	1.2 (0.5–3.1)
100,000–999,999	11.2 (2.3–55.3)†	2.4 (0.9–6.3)
≥1,000,000	13.8 (2.6–72.7)†	2.4 (0.8–7.3)
Trend test	<0.0001	<0.0001

Adjustment for gender, age, habits of cigarette smoking and alcohol consumption, HBeAg status and baseline ALT level.
* $p<0.05$; † $p<0.01$.

Table 6. Association between Baseline HBV Viral Load and ALT Elevation Afterward with Stratification by BCP 1762/1764 Variant

Baseline HBV DNA level copies/mL	BCP wild type	BCP A1762T/G1764A
	Adjusted odds ratio (95% CI)	Adjusted odds ratio (95% CI)
<300 (undetectable)	1.0 (referent)	1.0 (referent)
300–9,999	0.6 (0.2–1.7)	0.4 (0.1–2.0)
10,000–99,999	2.4 (0.9–6.4)	1.2 (0.3–4.3)
100,000–999,999	3.7 (1.4–9.8)*	3.6 (1.0–13.5)
≥1,000,000	2.6 (0.8–7.9)	7.3 (1.8–30.2)†
Trend test	<0.0001	<0.0001

Adjustment for gender, age, habits of cigarette smoking and alcohol consumption, HBeAg status and baseline ALT level.
* $p<0.05$; † $p<0.01$.

Summary

- A total of 320 (37.5%) subjects had at least one ALT elevation (≥45 U/L) during follow-up
- Male gender, baseline serum ALT and HBV DNA levels were statistically significantly associated with future ALT elevation
- Serum HBV DNA level was the strongest predictor of ALT elevation during long-term follow-up
- HBV genotype, precore G1896A and BCP A1762T/G1764A mutations were not statistically associated with ALT elevation
- The association between serum HBV DNA level and ALT elevation still holds when stratifying by HBV genotype, precore and BCP mutations

Conclusions

- High viral load remained the most important risk factor for ALT elevation regardless of genotype, precore and BCP mutations
- Lowering serum HBV viral load might be the key to prevent future elevation of ALT level

References

1. Chen CF, Yang HI, Iloeje UH, et al. Changes in serum HBV DNA level using a trajectory model to predict the risk of HCC in chronic hepatitis B patients: the R.E.V.E.A.L-HBV study. 18th APASL meeeting, March 23-26, 2008.

Disclosures

Uchenna Iloeje and Jun Su – Bristol-Myers Squibb employees.

The following people have nothing to disclose: Hwai-I Yang, Chin-Lan Jen, San-Lin You and Chien-Jen Chen.

Bristol-Myers Squibb